Icosabutate, a novel structurally engineered fatty-acid, exhibits potent anti-inflammatory and anti-fibrotic effects in a dietary mouse model resembling progressive human NASH

David A Fraser [2], Xiaoyu Wang[1], Cristina Alonso[3], Serene Lek[4], Tore Skjaeret[2], Detlef Schuppan[1,5] Institute of Translational Immunology, University Medical Center, Mainz, Germany; [2]NorthSea Therapeutics, 1411 DC Naarden, The Netherlands; [3]OWL Metabolomics, Parque Tecnológico de Bizkaia, Derio, Spain, [4]Clinnovate Health UK Ltd, Glasgow, UK, [5] Division of Gastroenterology, Beth Israel Deaconess Medical Center, Boston, MA, USA.

Disclosure: D. Schuppan acts as a consultant for NorthSea Therapeutics

Introduction

The liver harbours numerous fatty acid signalling-pathways and receptors of pivotal relevance to non-alcoholic steatohepatitis (NASH). Icosabutate ($C_{24}H_{38}O_3$) is a structurally engineered liver-targeted eicosapentaenoic acid (EPA) derivative designed to promote efficient targeting of metabolic, inflammatory and fibrotic pathways in the liver via resistance to both β-oxidation and esterification into complex lipids. We tested the effects of delayed therapy with icosabutate on hepatic inflammation and fibrosis in a dietary mouse model (CDAA) that reflects most facets of progressive human NASH.

Methods

Delayed treatment effects [introduced during final 6 weeks of a 12-week choline deficient (CD) L-amino acid diet (31% fat) in 9-week old C56BL/6J mice] of low (56mg/kg, ICOSA-L) and high (112mg/kg, ICOSA-H) dose icosabutate delivered orally or Bydureon (BY), a long-acting GLP-1 receptor agonist (0.4mg/kg weekly injected subcutaneously), on multiple indices of hepatic inflammation and fibrosis were assessed (9 mice per group). Unstained 5um thick FFPE liver histological sections were imaged using Genesis 200® Imaging System (HistoIndex Pte Ltd), a nonlinear multiphoton optical imaging system to detect changes in collagen fiber number per area, fiber length and thickness. Changes in hepatic lipidomic profiles were also assessed to compare icosabutate with an equimolar (to ICOSA-H) dose of EPA.

Results: Bodyweight & hepatic inflammation

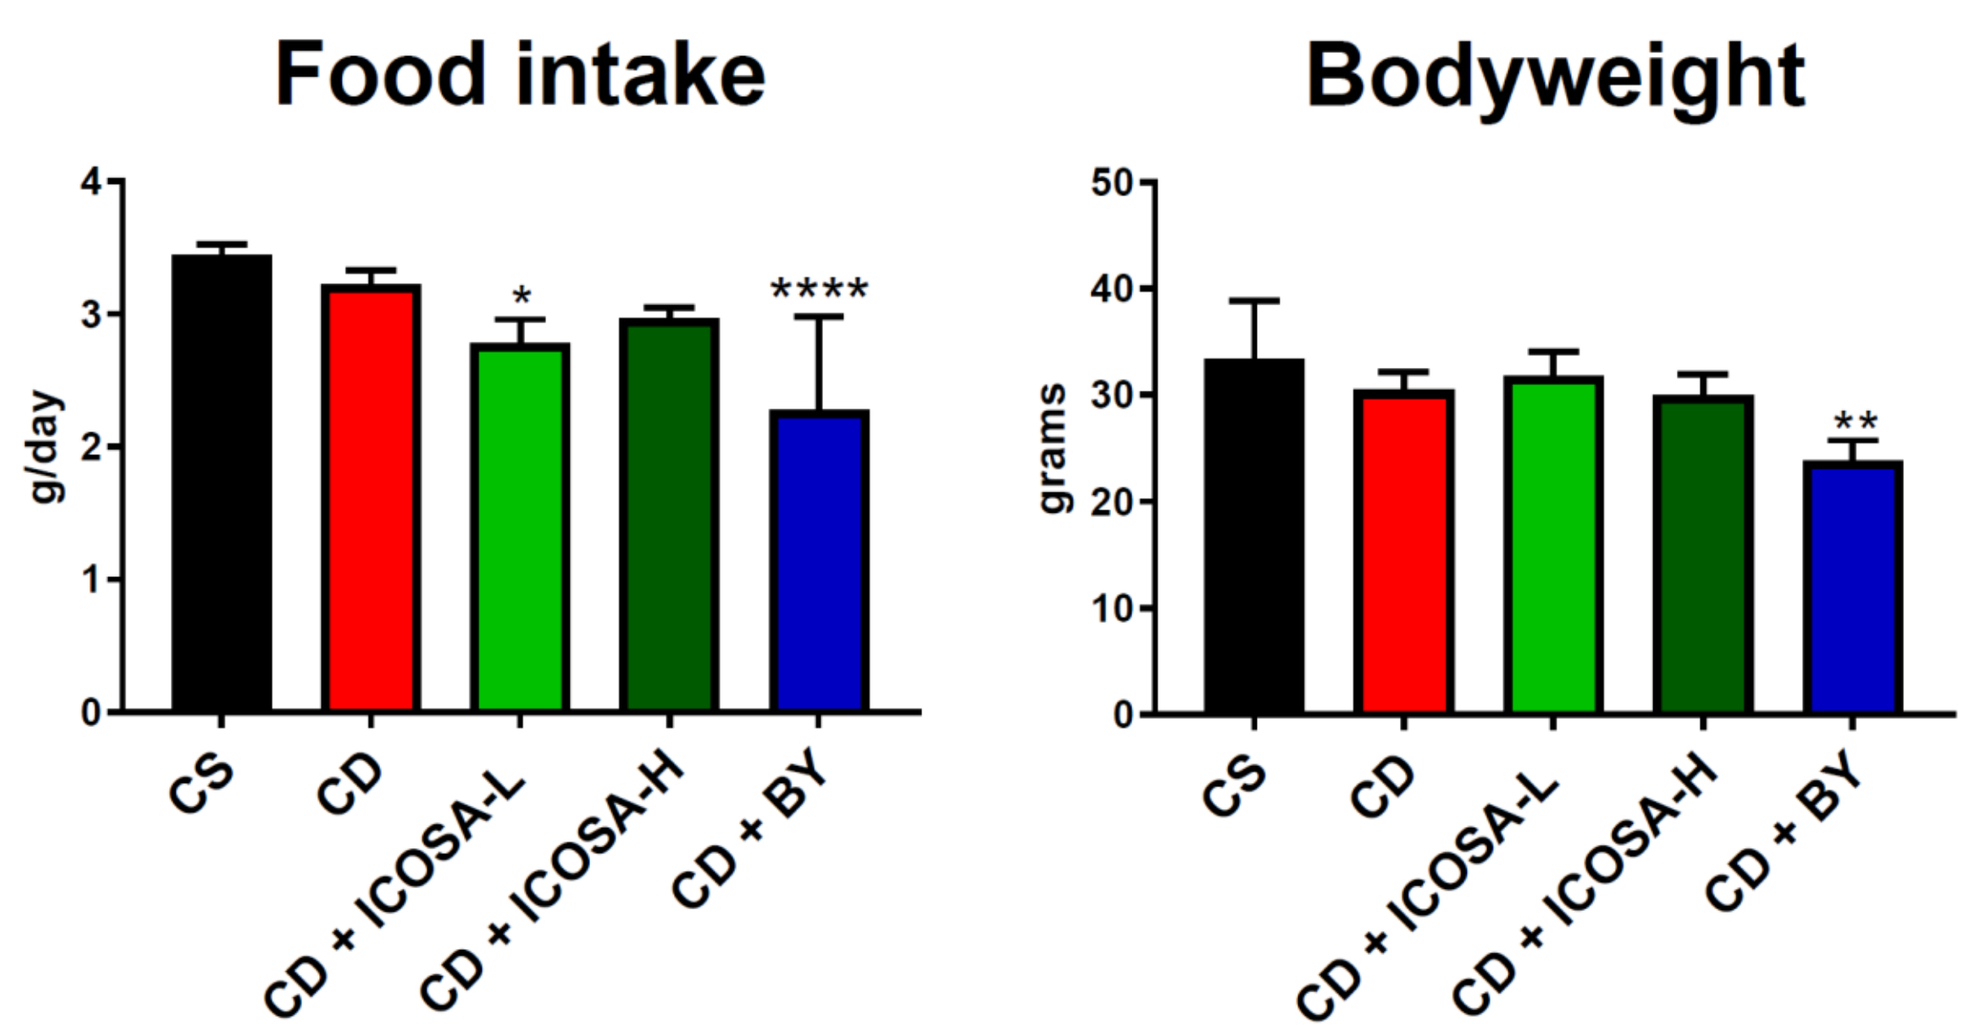
Food intake
g/day
Bodyweight
grams
CS
CD
CD + ICOSA-L
CD + ICOSA-H
CD + BY

Left: Treatment with the GLP-1R agonist, Bydureon, but not icosabutate, is associated with a significant decrease in body weight.**p<0.01, ****p<0.0001 vs CD

Below: Despite maintaining normal bodyweight, icosabutate reduces hepatic transcripts of inflammatory genes at least as efficiently as Bydureon. Neither agent decreases hepatic macrophage (CD68) cell content. *p<0.05, **p<0.01, ***p<0.001 vs CD

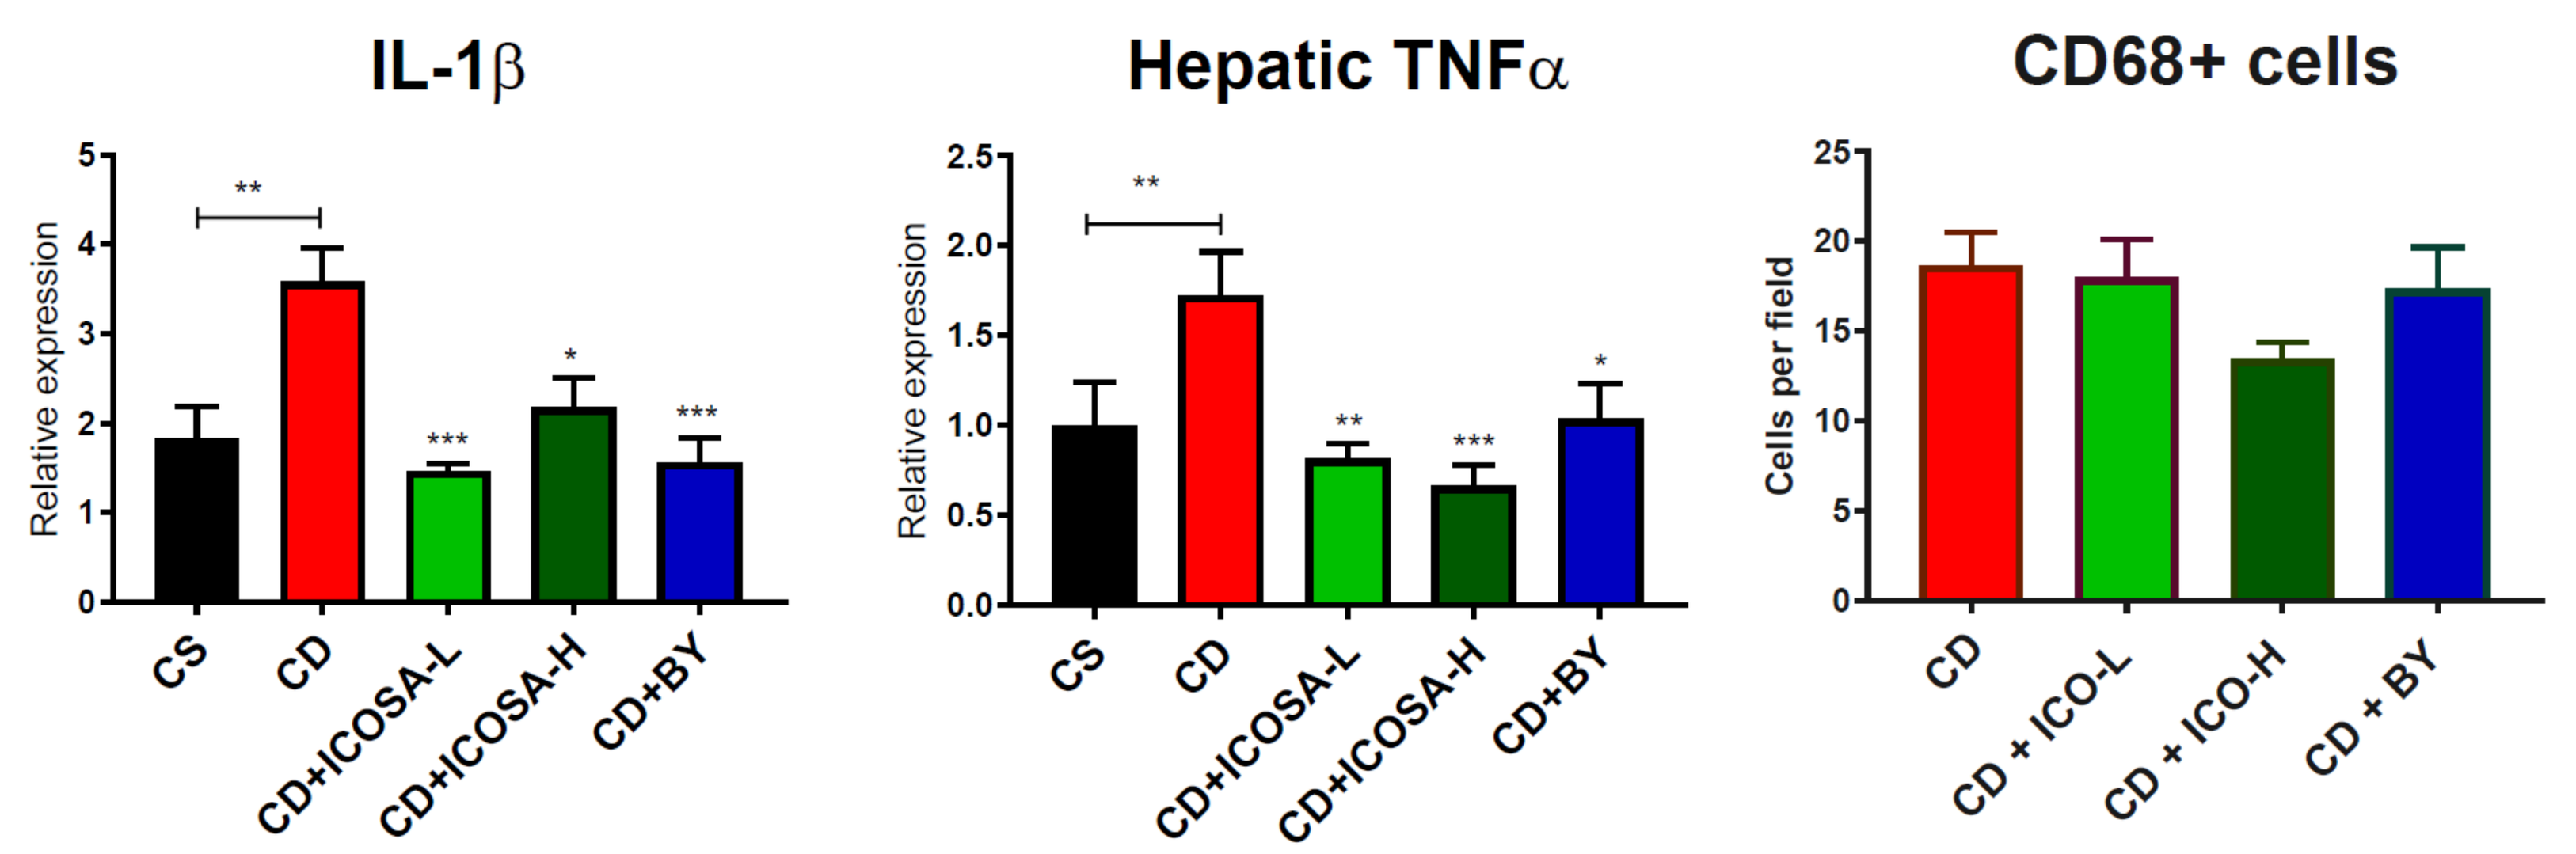
IL-1β
Relative expression
Hepatic TNFα
Relative expression
CD68+ cells
Cells per field
CS
CD
CD+ICOSA-L
CD+ICOSA-H
CD+BY
CD + ICO-L
CD + ICO-H
CD + BY

Hepatic fibrosis

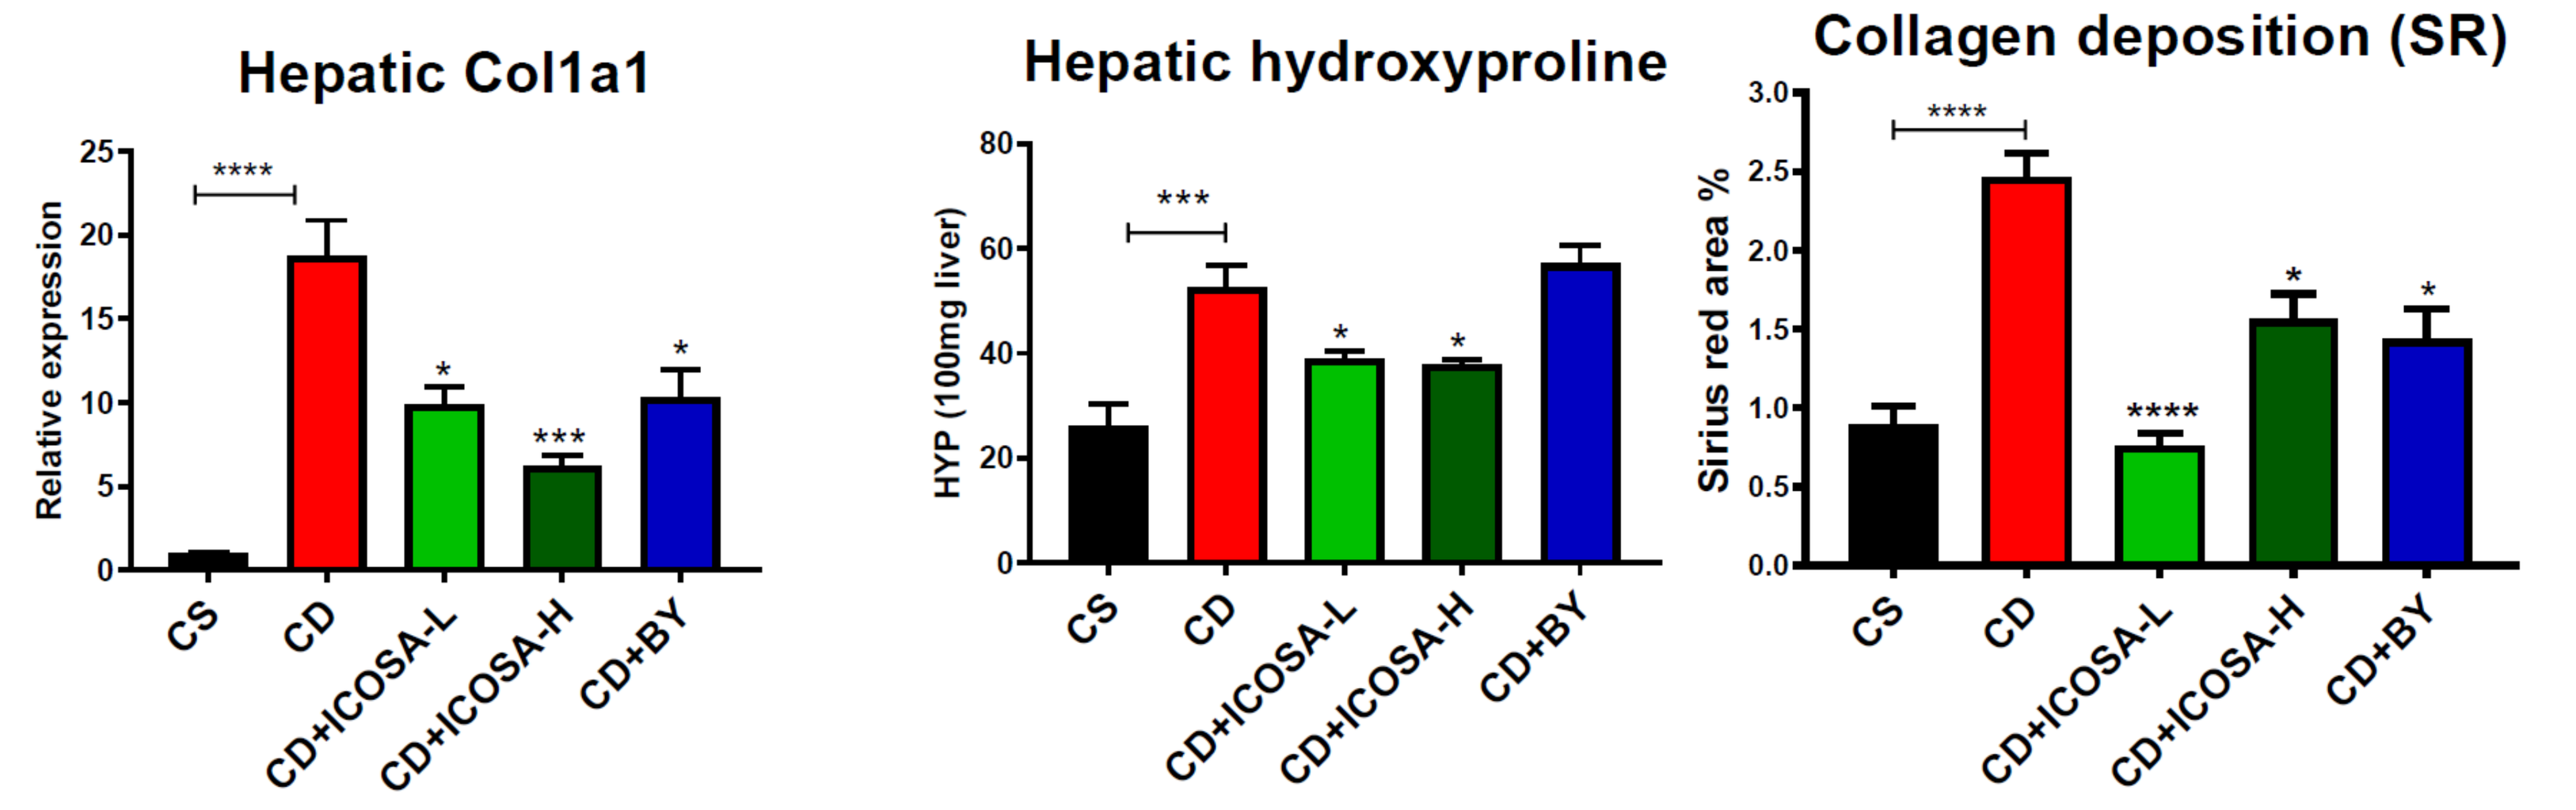
Hepatic Col1a1
Relative expression
Hepatic hydroxyproline
HYP (100mg liver)
Collagen deposition (SR)
Sirius red area %
CS
CD
CD+ICOSA-L
CD+ICOSA-H
CD+BY

Above: Delayed treatment with icosabutate (both doses) decreases hepatic fibrosis as measured via Col1A1 gene expression, hydroxyproline content and Sirius Red (SR) morphometry. Bydureon also reduces Col1A1 gene expression and collagen deposition but was without effect on hydroxyproline (no effects were seen with EPA, data not shown). *p<0.05, ***p<0.001, ****p<0.0001 vs CD

Right: Sirius Red stain: representative liver sections from individual mice in each treatment group

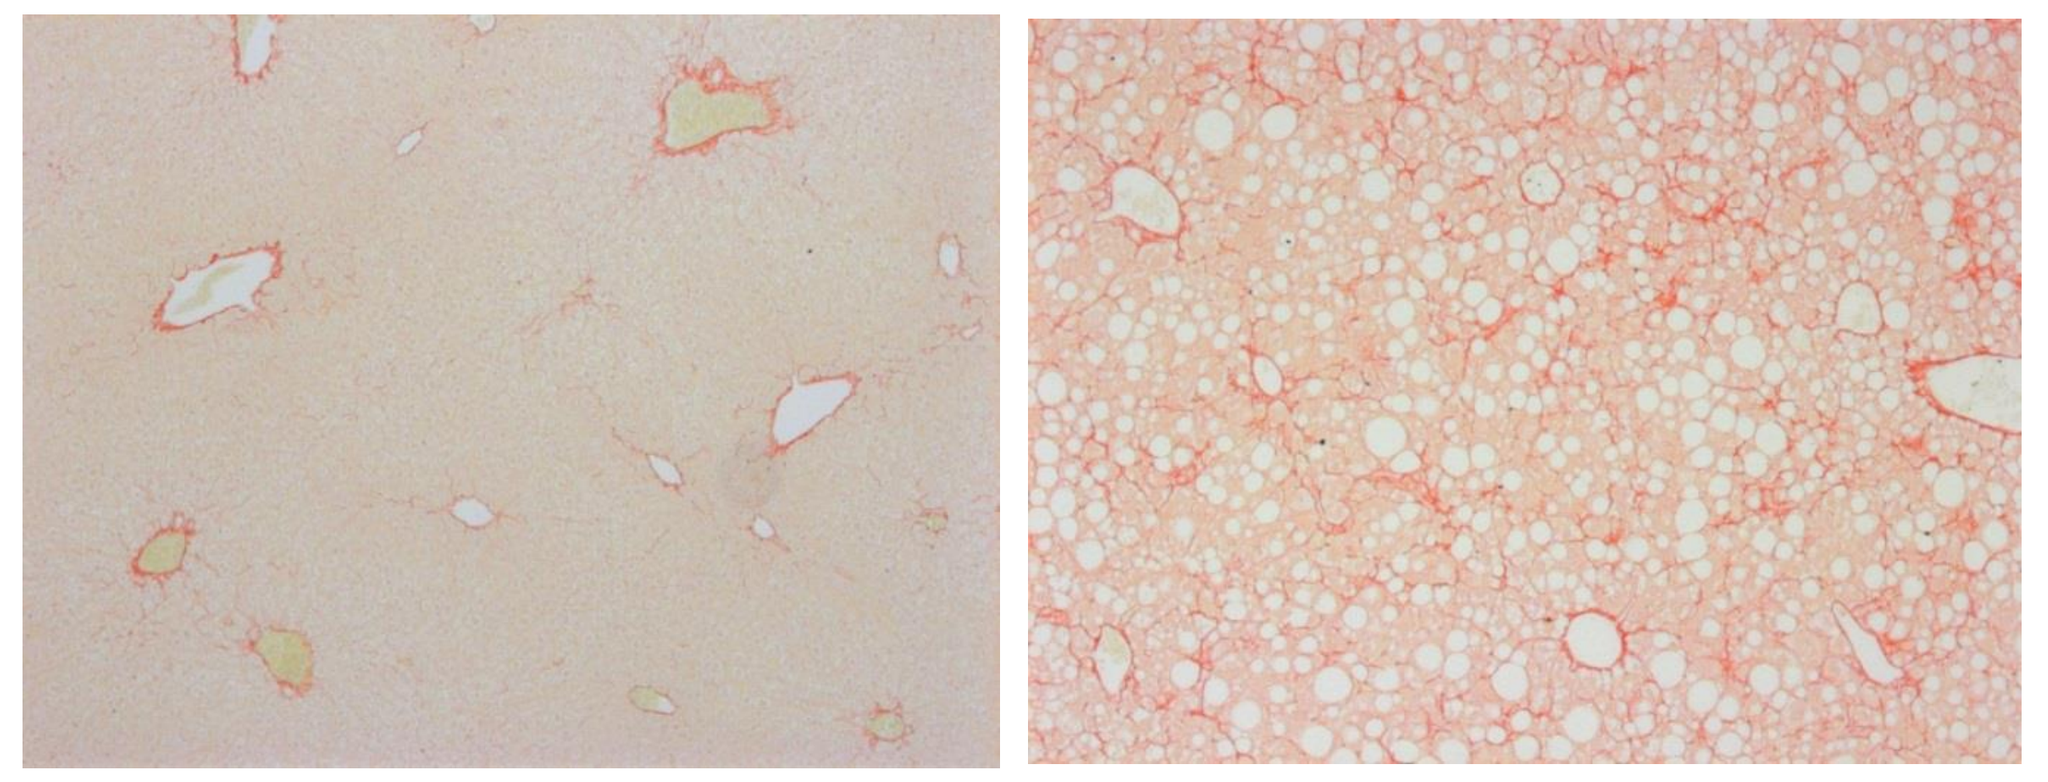

Choline sufficient (CS) Choline deficient (CD)

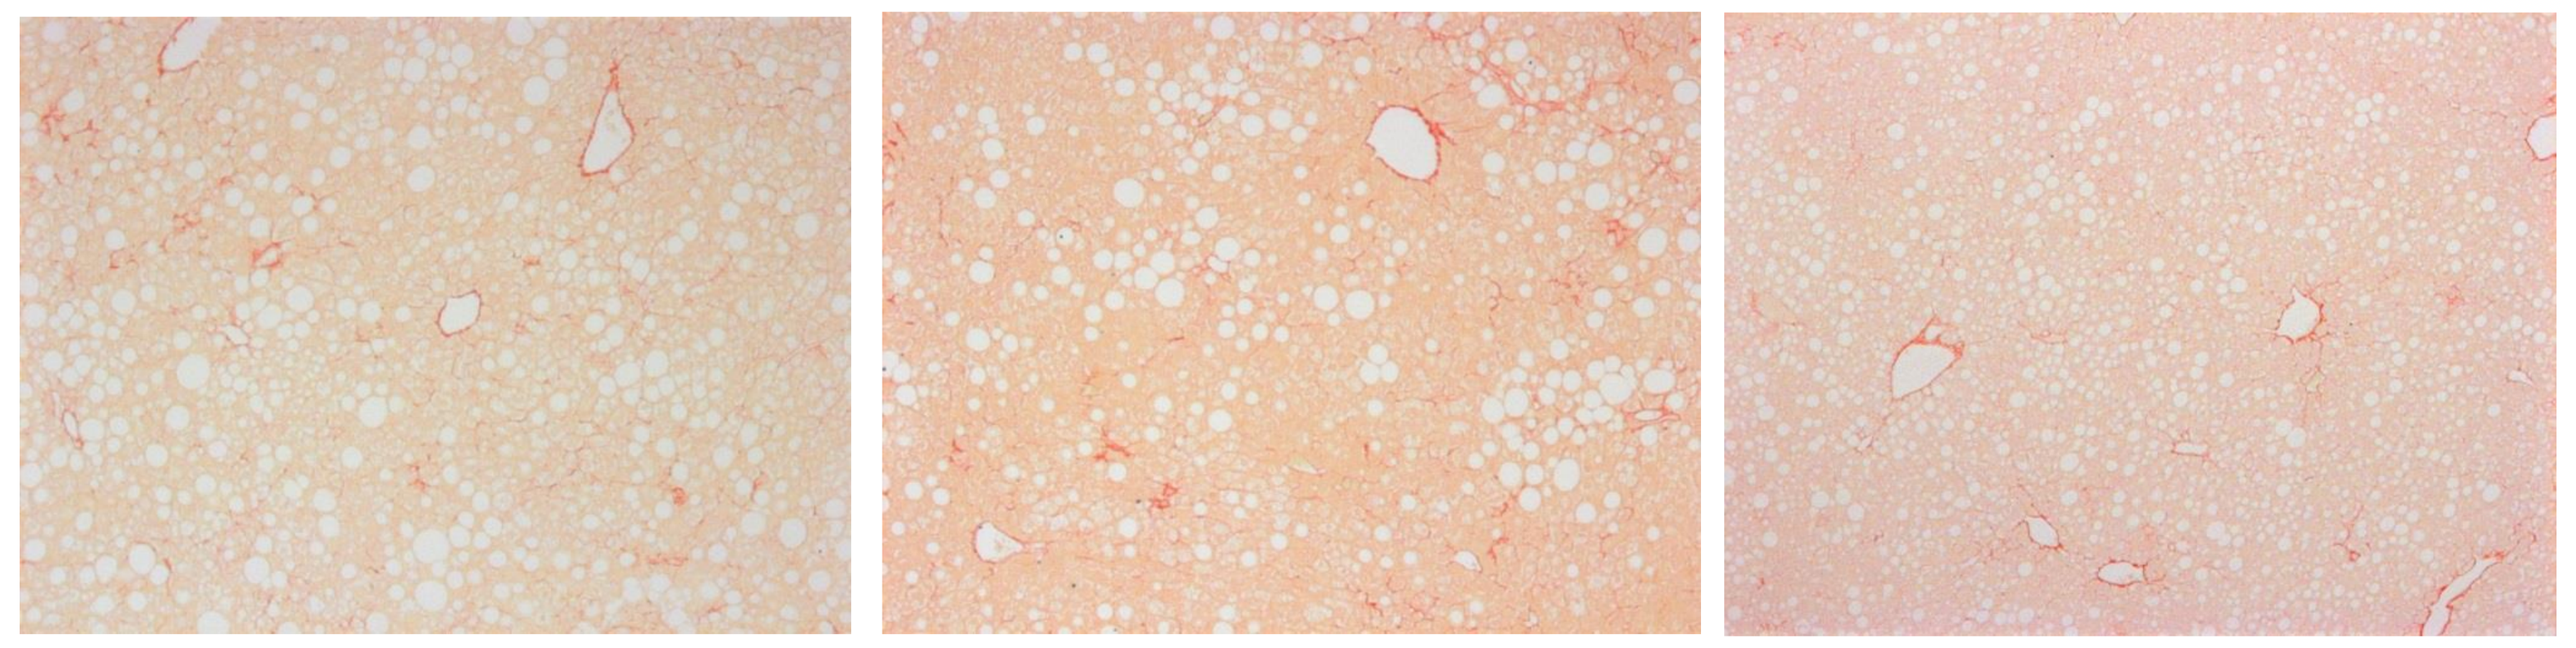

CD + ICOSA-L CD + ICOSA-H CD + BY

Below: Icosabutate (only ICOSA-L liver samples tested) significantly reduces collagen fiber number by 51% which reflects the almost complete disappearance of short (<8.5um) and thin (<3.5um) fibers. No significant effect was seen with Bydureon treatment. **p=0.01

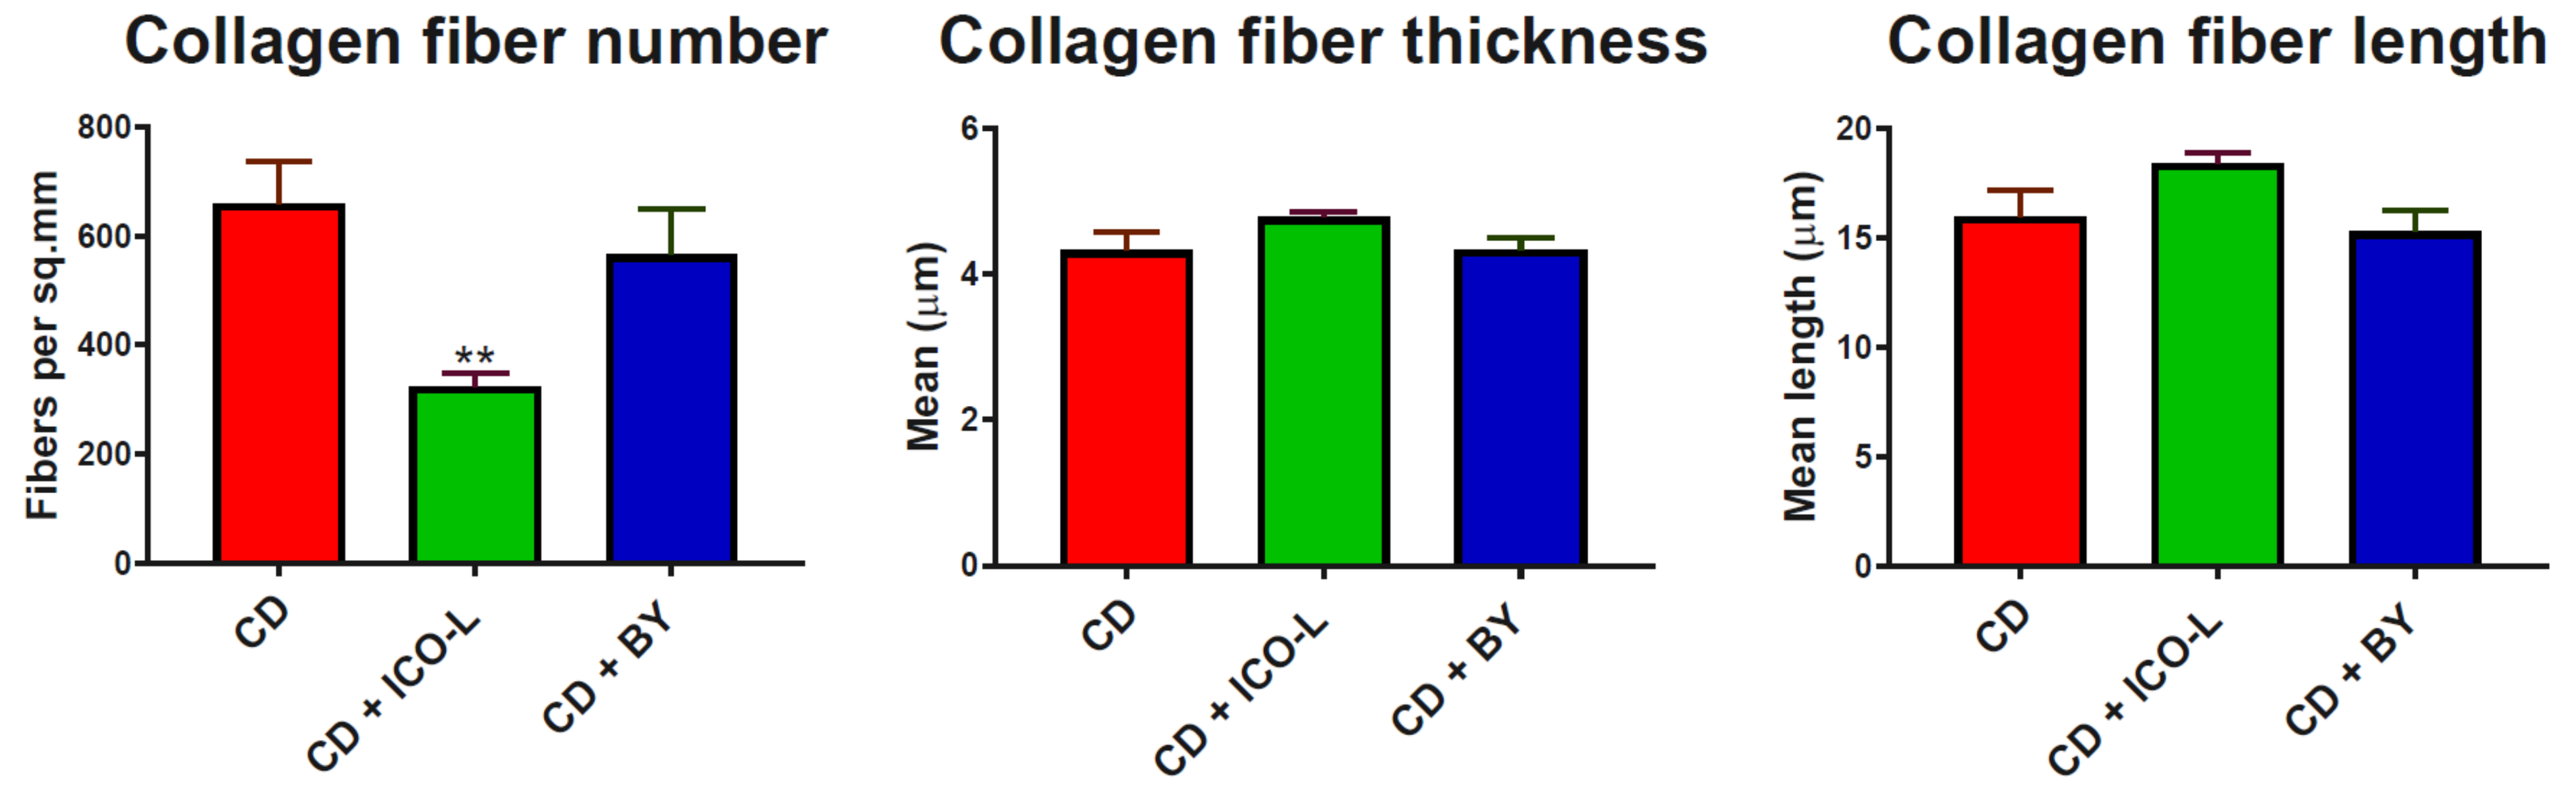
Collagen fiber number
Fibers per sq.mm
Collagen fiber thickness
Mean (µm)
Collagen fiber length
Mean length (µm)
CD
CD + ICO-L
CD + BY

Hepatic lipidomics

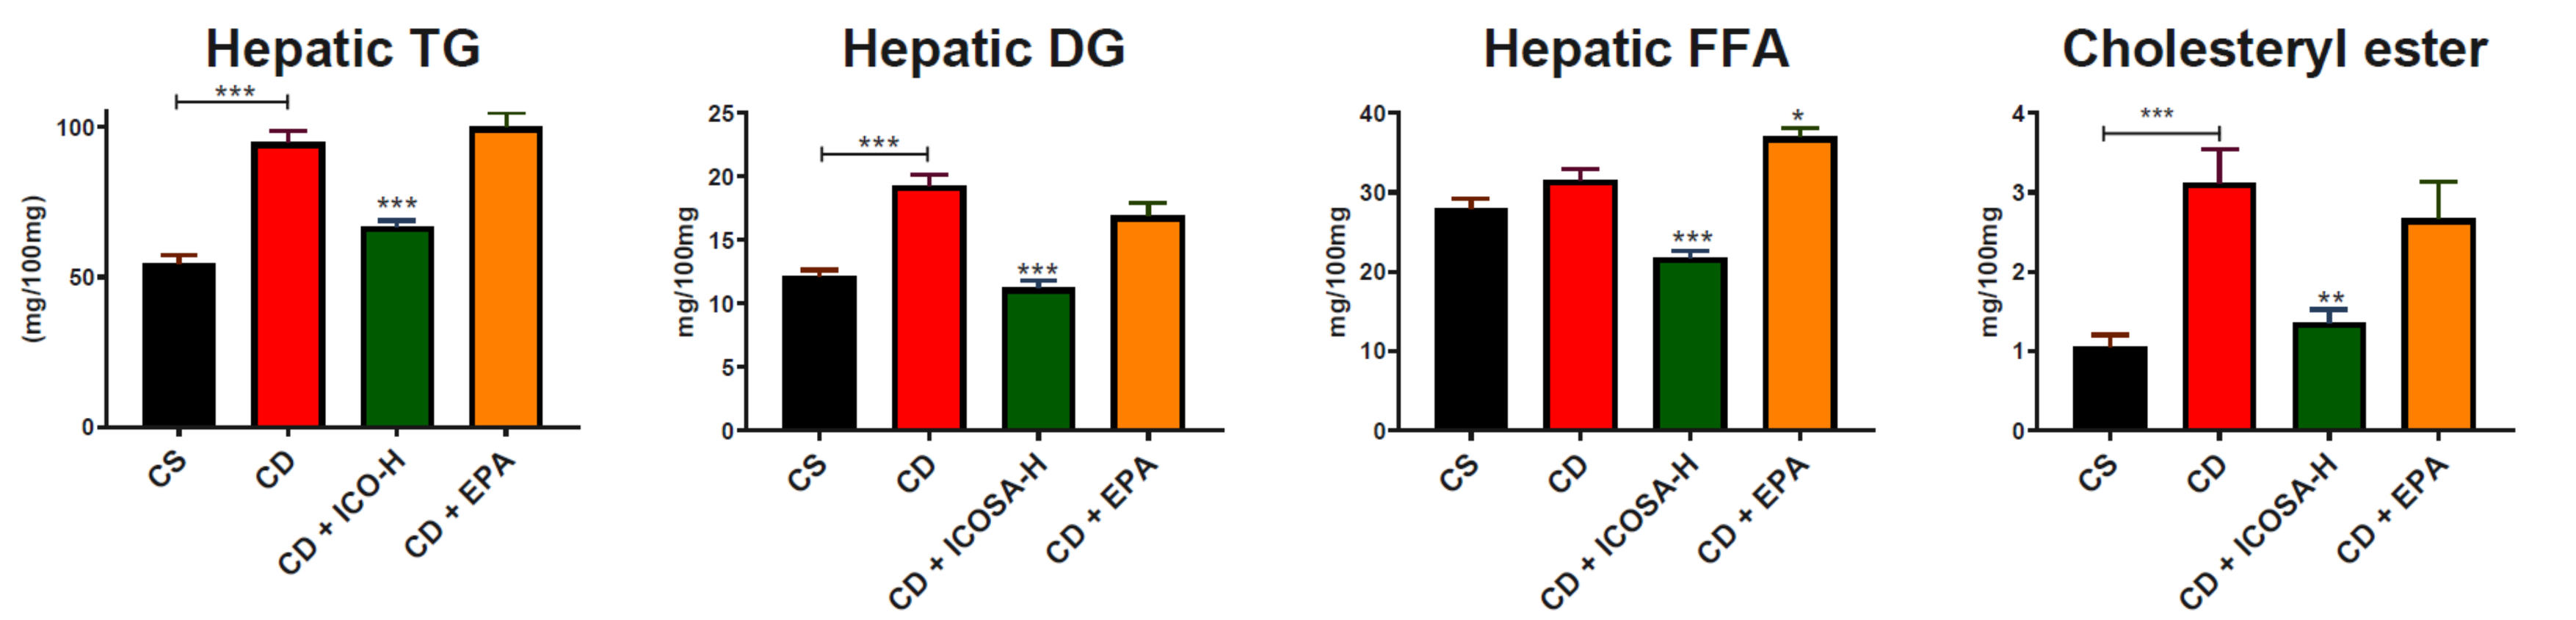
Hepatic TG
(mg/100mg)
Hepatic DG
mg/100mg
Hepatic FFA
mg/100mg
Cholesteryl ester
mg/100mg
CS
CD
CD + ICO-H
CD + ICOSA-H
CD + EPA

Above: Icosabutate, but not structurally unmodified EPA, reverses CD diet induced changes in hepatic concentrations of diacylglycerol (DG), TG and CE along with significant reductions in FFA concentration. *p<0.05, **p<0.01, ***p<0.001 vs CD.
Below: Icosabutate, but not EPA, has pronounced effects on concentrations of NASH-associated oxidised omega-6 lipid metabolites.

	CD-EPA *vs*. CD		CD-ICOSA-L *vs*. CD		CD-ICOSA-H *vs*. CD	
Class	$\log_2$ (fold-change)	Student´s t-test (p-value)	$\log_2$ (fold-change)	Student´s t-test (p-value)	$\log_2$ (fold-change)	Student´s t-test (p-value)
HETE	-0.11	7.77E-01	-0.83	2.45E-02	-1.45	2.27E-03
HODE	0.40	9.07E-02	-0.48	4.97E-02	-0.62	1.34E-02
DiHOME	0.31	1.80E-01	-0.42	3.88E-02	-0.52	7.60E-03
TriHOME	0.46	1.09E-01	-1.14	5.49E-04	-1.59	2.32E-04
oxoODE	-0.57	9.50E-02	0.90	2.87E-02	1.01	1.10E-03
OXLAMs	0.34	1.16E-01	-0.37	7.56E-02	-0.49	1.59E-02
Total oxidised FA	0.20	3.74E-01	-0.51	3.35E-02	-0.75	2.75E-03

≤-4	-4	-3	-2	-1	1	2	3	4	≥4

p<0.05 p<0.01 p<0.001

Conclusions

- **Oral treatment with icosabutate has a pronounced beneficial effect on fibrosis in a delayed intervention CDAA mouse NASH model**
- **At a human equivalent dose of approx. 300mg/day, icosabutate brought fibrosis levels back to baseline despite delayed onset of treatment**
- **The reduction in fibrosis was associated with a pronounced >50% reduction in collagen fiber number**
- **Hepatic lipidomic analysis identified markedly reduced hepatic TG, DAG, FFA and CE along with potent reductions in oxidised omega-6 fatty acid metabolites**
- **Icosabutate, at a human equivalent dose of 300-600mg/day, shows excellent potential for treating human NASH with potent effects upon hepatic lipotoxicity, inflammation and fibrosis**

CONTACT INFO: David A. Fraser, CSO, david.fraser@northseatherapeutics.com