A Phase 1 Study in Healthy Volunteers to Evaluate the Safety, Tolerability and Pharmacokinetics of SEFA-6179, a Fully-Synthetic Medium Chain Fatty Acid Analogue in Clinical Development for Intestinal Failure-Associated Liver Disease

Carine Beysen[1], David A Fraser[1], Tore Skjæret[1], Emilie Lasson[1], Celeste R. MacElrevey[2], Jim Bush MBChB[2] and Stephen Harrison[1]

[1]NorthSea Therapeutics, Amsterdam, The Netherlands and [2]Labcorp Clinical Research Unit Limited, Leeds, United Kingdom.

INTRODUCTION

- Patient with intestinal failure who rely on parental nutrition (PN) are at risk for developing Intestinal Failure-Associated Liver Disease (IFALD)
- IFALD is a multifactorial liver condition characterized by the development of hepatic steatosis, inflammation, cholestasis and fibrosis.
- There are no approved drug therapies available, and treatment primarily involves optimizing nutrition support to minimize potential liver injury.
- SEFA-6179, also known as NST-6179, is a Structurally Engineered Medium Chain Fatty Acid (MC-SEFA) analogue of decanoic acid and is currently in clinical development for the treatment of IFALD

DECANOIC ACID

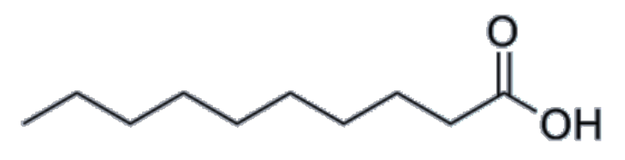
O
OH

SEFA-6179

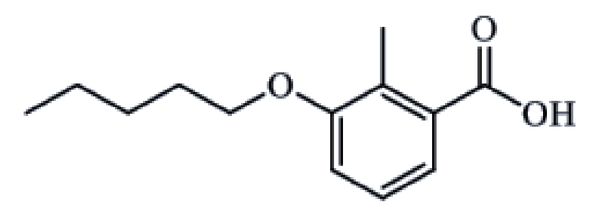
O
O
OH

- SEFA-6179 is designed to maintain the passive transport from the gut to the liver and cellular uptake that is characteristic of natural MCFAs, whilst offering enhanced pharmacological effects via avoidance of rapid metabolism.
- SEFA-6179 targets multiple receptors (both G-protein coupled and nuclear) that independently regulate pivotal pathways involved in IFALD pathophysiology (see figure below).

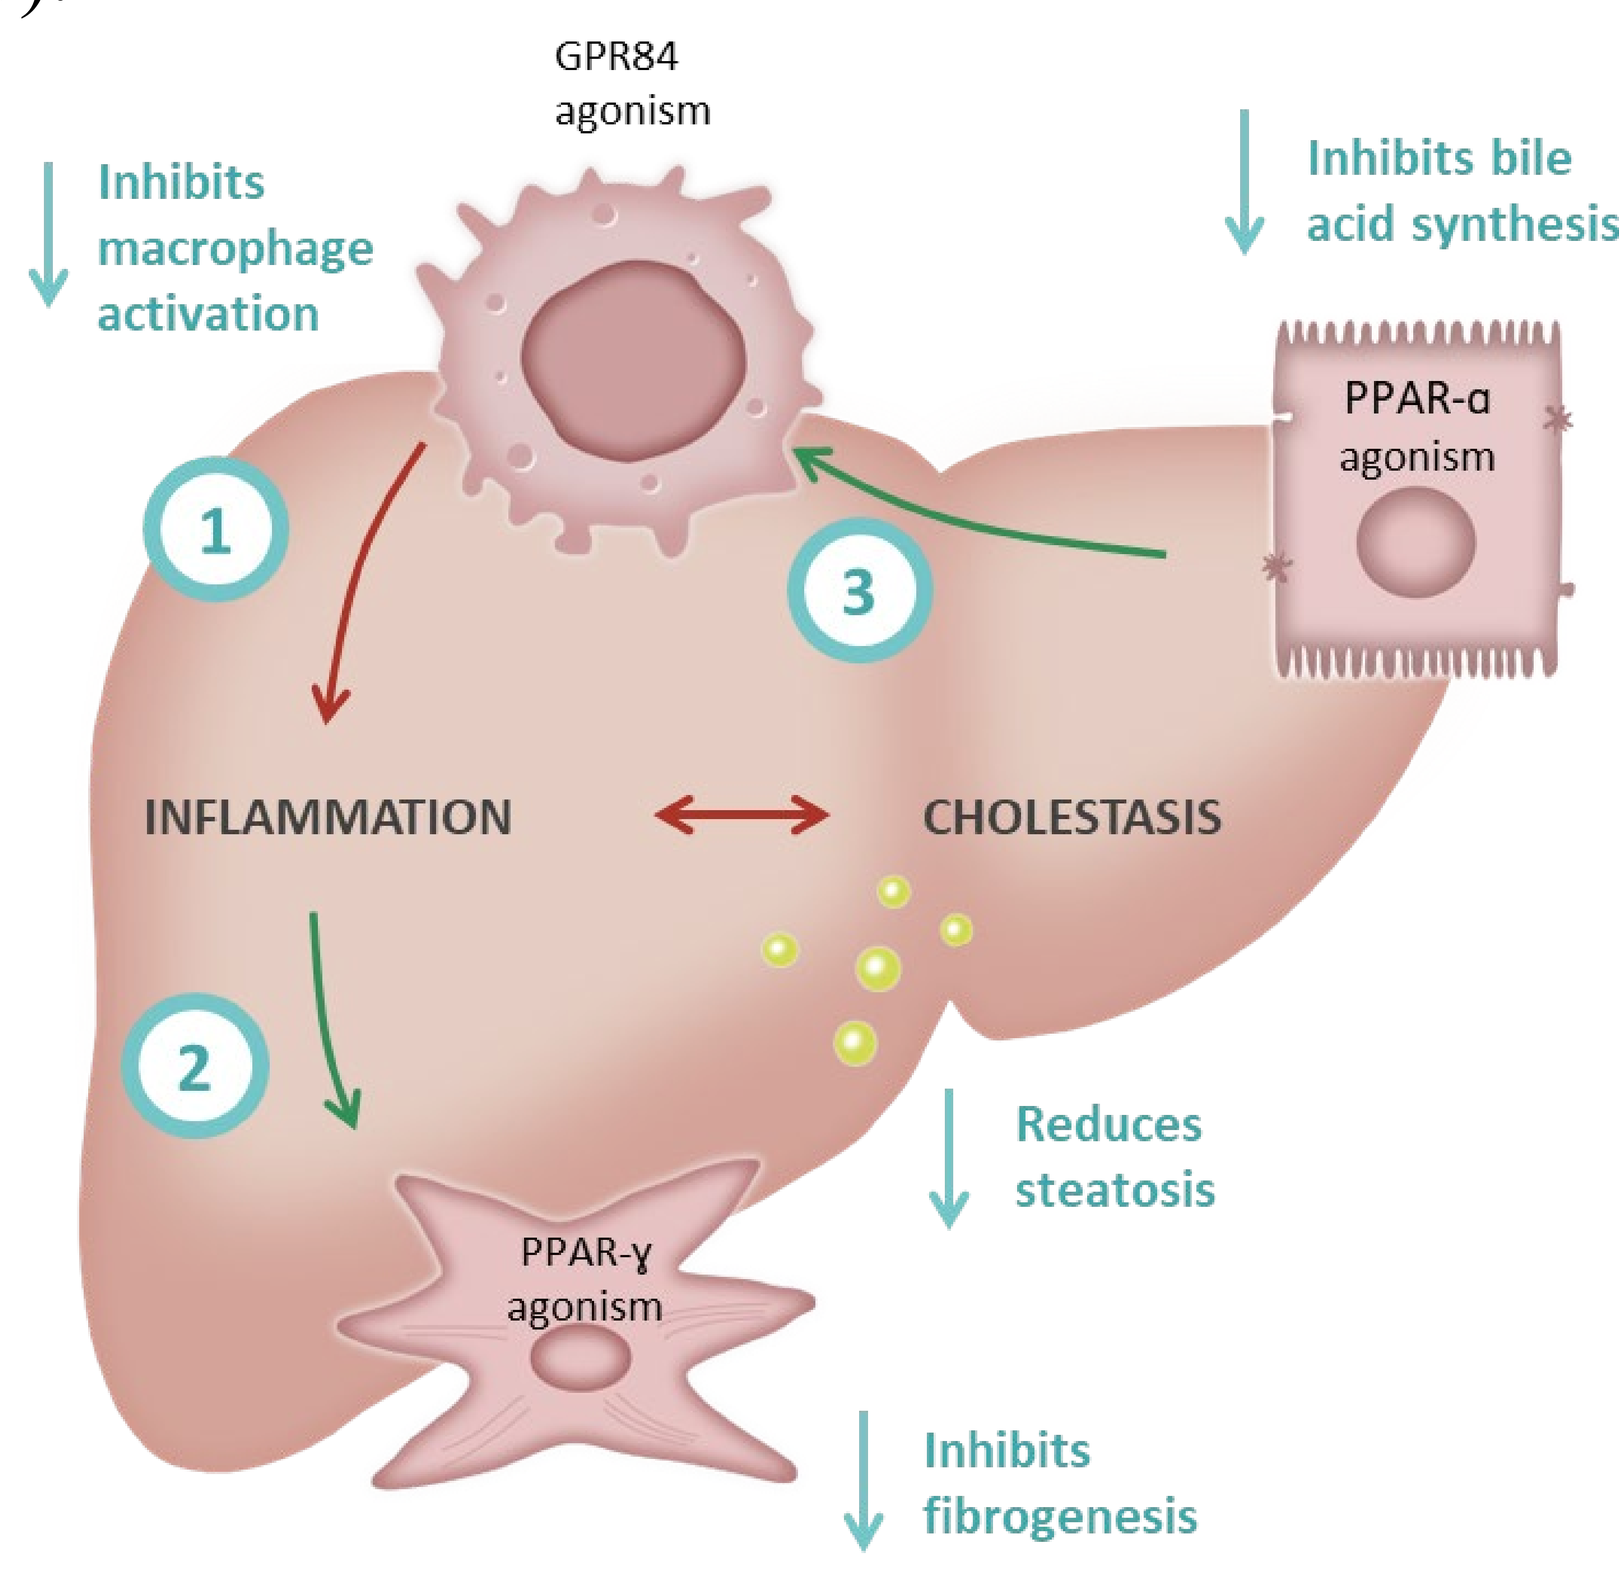
GPR84
agonism
Inhibits
macrophage
activation
Inhibits bile
acid synthesis
PPAR-α
agonism
1
3
INFLAMMATION
CHOLESTASIS
2
Reduces
steatosis
PPAR-γ
agonism
Inhibits
fibrogenesis

STUDY OBJECTIVES

To assess the safety, tolerability and pharmacokinetics of single and multiple oral doses of SEFA-6179 in healthy volunteers to establish safe dose regimens for future studies in patients.

STUDY DESIGN

- This was a double-blind randomized, placebo-controlled phase 1 study conducted in the United Kingdom in healthy male and female adults
- Single (Groups A1-5) and multiple (Groups B1-3) ascending QD doses of SEFA-6179 were tested

Part A:

Group A1: 50 mg SEFA-6179 (n=6) or Placebo (n=2) → Group A2: 200 mg SEFA-6179 (n=6) or Placebo (n=2) → Group A3: 400 mg SEFA-6179 (n=6) or Placebo (n=2) → Group A4: 600 mg SEFA-6179 (n=6) or Placebo (n=2) → Group A5: 1000 mg SEFA-6179 (n=6) or Placebo (n=2)

Part B:

Group B1: 200 mg SEFA-6179 (n=8) or Placebo (n=2) QD, 14 Days → Group B2: 400 mg SEFA-6179 (n=8) or Placebo (n=2) QD, 14 Days → Group B3: 1000 mg SEFA-6179 (n=8) or Placebo (n=2) QD, 14 Days

- All doses were administered orally as a solid in hard gelatin capsules after an overnight fast of at least 10 hours
- Adverse events (AEs), vital signs, electrocardiographic (ECG) findings, physical examinations, clinical laboratory values and PK of blood samples were assessed

RESULTS

❖ Baseline Characteristics

Part A (SAD)	Placebo	50 mg	200 mg	400 mg	600 mg	1000 mg
Subjects (n)	10	6	6	6	6	6
Age (years)	45.7 (14.5)	41.7 (13.1)	40.0 (13.9)	41.7 (16.0)	37.3 (14.7)	45.7 (15.9)
Sex (male/female)	6/4	3/3	1/5	3/3	4/2	4/2
Body Weight (kg)	76.6 (12.5)	74.2 (12.9)	69.5 (14.3)	68.3 (14.3)	82.4 (15.1)	78.5 (10.6)
Body Mass Index (kg/m²)	25.6 (2.6)	25.2 (4.3)	24.6 (3.6)	23.9 (4.4)	27.4 (3.8)	26.4 (2.1)

Part B (MAD)	Placebo	200 mg	400 mg	1000 mg
Subjects (n)	6	8	8	8
Age (years)	40.8 (15.5)	40.5 (15.3)	45.1 (10.2)	50.6 (9.8)
Sex (male/female)	3/3	5/3	4/4	8/0
Body Weight (kg)	80.9 (14.6)	76.0 (14.2)	78.2 (17.7)	81.6 (10.4)
Body Mass Index (kg/m²)	26.5 (2.7)	25.5 (2.8)	26.1 (3.9)	27.4 (3.5)

Data are mean (SD)

❖ Treatment-Emergent Adverse Events (TEAE)

Part A (SAD)	Placebo	50 mg	200 mg	400 mg	600 mg	1000 mg
Subjects (n)	10	6	6	6	6	6
TEAEs						
Overall	4 (40%)	2 (33%)	2 (33%)	2 (33%)	2 (33%)	2 (33%)
Mild	4 (40%)	2 (33%)	2 (33%)	2 (33%)	2 (33%)	1 (16.5%)
Moderate	---	---	---	---	---	1 (16.5%)
Severe	---	---	---	---	---	---
Treatment-related TEAEs						
Overall	---	---	---	---	---	1 (16.5%)
Mild	---	---	---	---	---	---
Moderate	---	---	---	---	---	1 (16.5%)
Severe	---	---	---	---	---	---
TEAE reported in > 1 subject per group						
Nervous system disorders						
Headache	---	2 (33%)	---	---	---	1 (16.5%)
Treatment -related TEAE						
Nervous system disorders						
Headache	---	---	---	---	---	1 (16.5%)

Data are number of subjects (% of subjects) with TEAE

❖ Treatment-Emergent Adverse Events *continued*

Part B (MAD)	Placebo	200 mg	400 mg	1000 mg
Subjects (n)	6	8	8	8
TEAEs				
Overall	2 (33.3%)	5 (62.5%)	6 (75.0%)	5 (62.5%)
Mild	2 (33.3%)	5 (62.5%)	5 (62.5%)	5 (62.5%)
Moderate	1 (16.7%)	2 (25.0%)	3 (37.5%)	1 (12.5%)
Severe	---	---	---	---
Treatment-related TEAEs				
Overall	2 (33.3%)	2 (25.0%)	5 (62.5%)	5 (62.5%)
Mild	2 (33.3%)	2 (25.0%)	4 (50.0%)	4 (50.0%)
Moderate	1 (16.7%)	---	3 (37.5%)	1 (12.5%)
Severe	---	---	---	---
TEAE reported in > 1 Subject per group				
Nervous system disorders				
Headache	1 (16.7%)	3 (37.5%)	5 (62.5%)	1 (12.5%)
Gastrointestinal disorders				
Abdominal pain upper	1 (16.7%)	2 (25.0%)	---	---
Constipation	---	2 (25.0%)	---	---
Nausea	1 (16.7%)	---	2 (25.0%)	---
Musculoskeletal and connective tissue disorders				
Back pain	---	---	---	4 (50.0%)
Limb discomfort	---	---	---	2 (25.0%)
Treatment -related TEAE in > 1 subject per group				
Nervous system disorders				
Headache	1 (16.7%)	---	5 (62.5%)	---
Musculoskeletal and connective tissue disorders				
Back pain	---	---	---	4 (50.0%)
Limb discomfort	---	---	---	2 (25.0%)

Data are number of subjects (% of subjects) with TEAE

❖ Pharmacokinetics

Part A (SAD)	50 mg	200 mg	400 mg	600 mg	1000 mg
C_{max} (ng/mL)	1100 (54.7)	4880 (49.9)	13200 (29.2)	18300 (33.7)	27200 (33.7)
T_{max} (h)*	1.49 (1-4)	1.75 (1-4)	2 (1.5-3)	1.5 (0.5-2)	2 (1-3)
$T_{1/2}$ (h)	0.845 (8.7)	1.05 (59.2)	1.38 (40.4)	1.71 (50.2)	2.91 (76.5)
AUC_{0-24} (h*ng/mL)	2680 (16.4)	10500 (28.6)	27200 (20.5)	39900 (22.8)	71700 (16.9)

Part B (MAD)	200 mg	400 mg	1000 mg
C_{max} (ng/mL)	6920 (28.0)	10000 (34.9)	31800 (39.9)
T_{max} (h)*	1.33 (1-2)	1.5 (1-3)	2 (1-2)
$T_{1/2}$ (h)	1.02 (17.0)	1.16 (14.5)	3.22 (20)
$AUC_{0-\tau}$ (h*ng/mL)	12800 (31.5)	24800 (26.8)	85700 (19.7)

Data are geometric mean (CV) or Median (minimum-maximum). Day 14 data are presented for Part B.*

❖ Treatment signals on liver enzymes (Group B3)

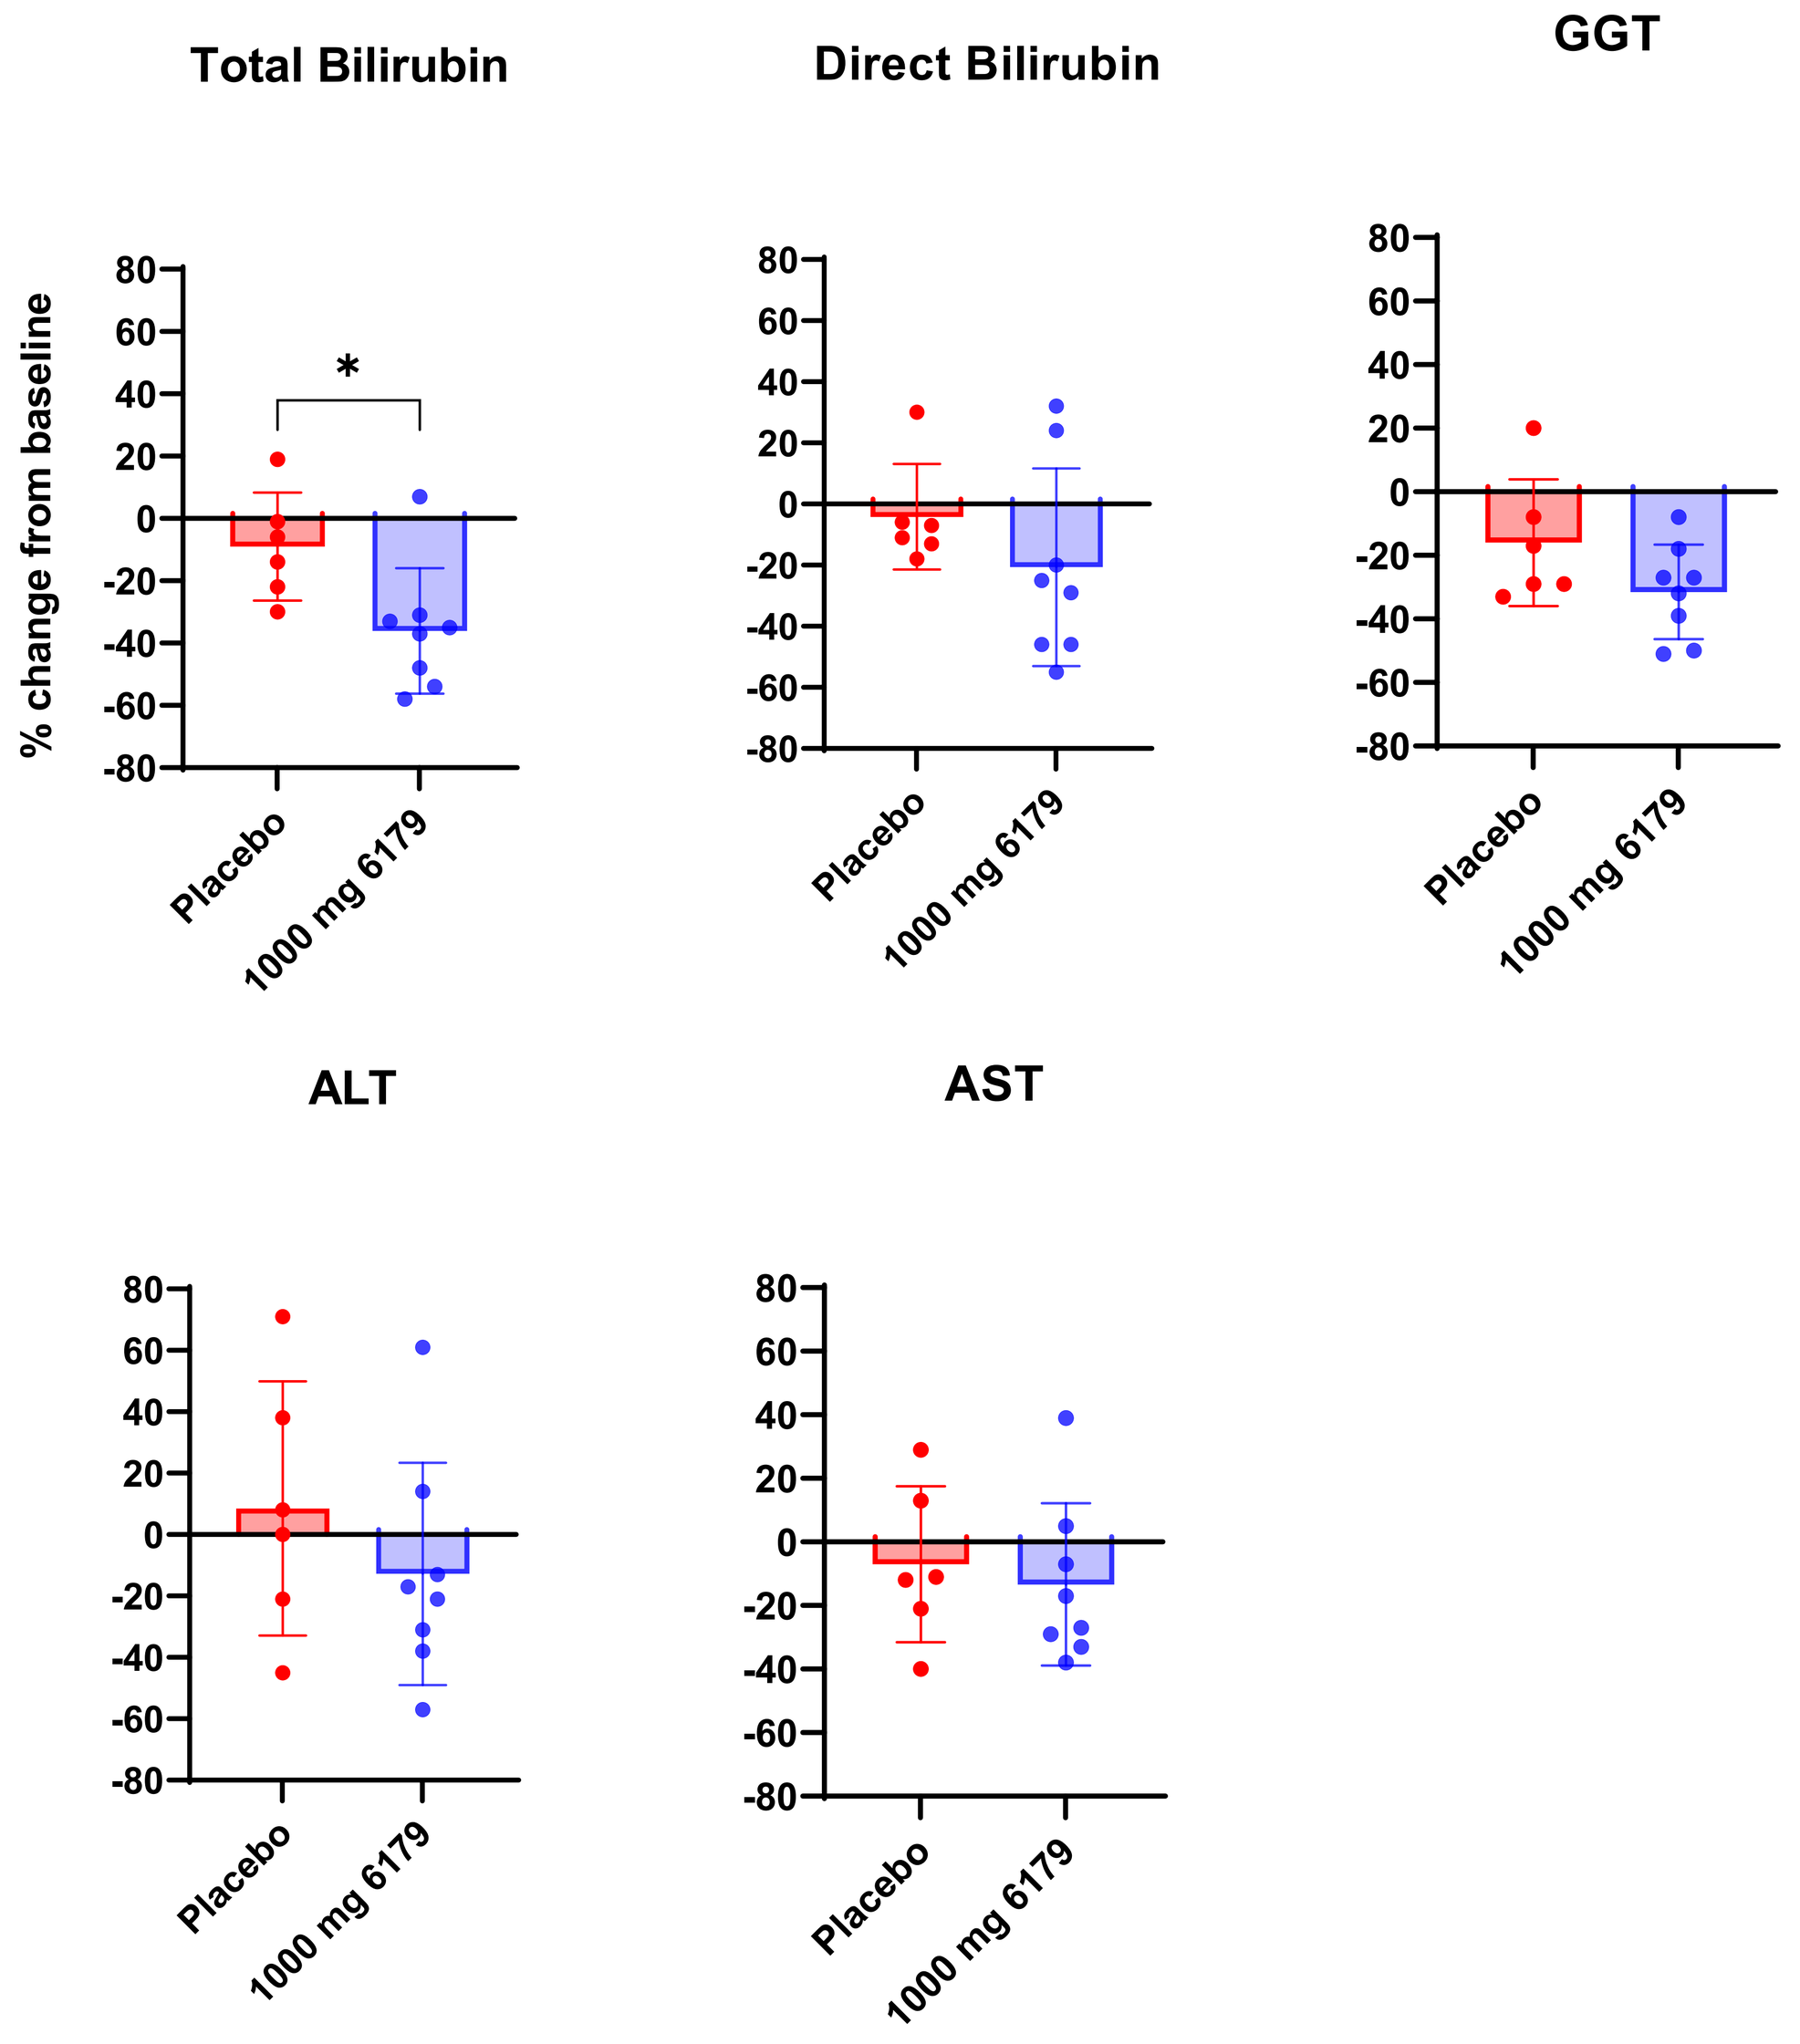
Total Bilirubin
Direct Bilirubin
GGT
ALT
AST
% change from baseline
*
Placebo
1000 mg 6179

Data are presented as scatter dot plots with mean and SD

DISCUSSION

- A total of 54 healthy male and female subjects were exposed to at least 1 dose of SEFA-6179
- QD doses up to 1000 mg were considered safe and well-tolerated and providing an adequate safety threshold for the subsequent phase 2a study.
- There were no treatment- or dose-related trends and no clinically significant findings in clinical laboratory evaluations, vital signs, 12-lead ECG data, or physical examination findings during the study
- The majority of TEAEs were mild and resolved without treatment
- There were no severe TEAEs or SAEs during the study
- No subjects discontinued the study due to a TEAE
- Headaches were the most common TEAE
- There were no apparent trends with occurrence of TEAEs with increasing dose of SEFA-6179
- SEFA-6179 appeared rapidly in plasma following a single dose, with a T_{max} between 1.25 and 2 hours and was eliminated with a $T_{1/2}$ between 1 to 3 hours
- There was minimal accumulation of SEFA-6179 after 14 days of dosing, with accumulation rations of 1.1 to 1.2 for AUC0-τ and C_{max}
- Urinary excretion of unchanged SEFA-6179 was minimal following single and multiple doses.
- Systemic exposure of SEFA-6179 generally increased in a dose proportional manner in terms of AUC and Cmax.
- Improvements in liver enzymes after 14 days of 1000 mg QD dosing may be an early signal of treatment efficacy.

CONCLUSIONS

SEFA-6179 is well tolerated and exhibits an acceptable safety and PK profile supporting further clinical development for the treatment of IFALD as well as potentially other cholestatic liver diseases. A phase 2a study in adult patients with IFALD is starting in H1, 2023.

ACKNOWLEDGEMENT

We would like to express our sincere gratitude to all the participants who generously gave their time to participate in this study.

RELATATED VARS AWARD ORAL PRESENTATION

Absorption of an Engineered Medium Chain Fatty Acid in Two Short Bowel Syndrome Minipig Models presented by Scott C. Fligor, Boston Children's Hospital, Boston, MA

--- Saturday, April 22 • 10:30 AM – 12:30 PM PT ---

Disclosure and contact: This research was funded by NorthSea Therapeutics, carine.beysen@northseatherapeutics.com

RESEARCH POSTER PRESENTATION DESIGN © 2019
www.PosterPresentations.com